LC-MS/MS Validation of a Novel 3'3'-cGAMP Monoclonal ELISA

Julianna Blaylock†, Kerry Metcalfe†, Kaylee Wilburn‡, Chris Waters‡, and Daniel Tew†

†Cayman Chemical Company, Ann Arbor, MI; ‡Department of Microbiology & Molecular Genetics, Michigan State University, East Lansing, MI

KEY FINDING A highly sensitive and specific ELISA to 3'3'-cGAMP that correlates well with LC-MS/MS.

INTRODUCTION

Second messengers are small molecules that cause a wide variety of cellular changes transcriptionally, translationally, and post-translationally. 3'3'-cGAMP is a bacterial second messenger produced from ATP and GTP by specific dinucleotide cyclases.[1] It is produced and regulated *via* two different pathways, which use distinct classes of synthases, effectors, and phosphodiesterases (PDEs).[2,3] 3'3'-cGAMP binds to riboswitches to regulate motility, biofilm formation, virulence, and colonization through gene transcription.[4] The study of cyclic dinucleotides (CDNs) in bacterial innate immunity is a growing area of research. Current detection methods utilize mass spectrometry, which can be both costly and timely. In addition to that, the isolation and purification of CDNs can be complicated and tedious. Rapid and accurate detection methods are critical to enable researchers to study and/or identify the relevant biological pathways.

This poster will show LC-MS/MS validation of a novel 3'3'-cGAMP monoclonal enzyme-linked immunosorbent assay (ELISA). The data provided will demonstrate the ELISA is sensitive, specific, and in good correlation with LC-MS/MS.

METHODS

Our objective was to quantitate bacterial 3'3'-cGAMP levels by LC-MS/MS and ELISA. The protocol for sample preparation is shown in brief below and found in full at Wilburn *et al.* 2023.[5]

Bacterial Cultures

Overnight cultures of 3'3'-cGAMP-producing recombinant *E. coli* were back diluted 1:100 with 1 mM IPTG induction and grown until an OD_{600} of 0.1. CFUs were calculated, and phage were added at an MOI of 1. At different timepoints after phage were added, two 1 ml aliquots from each sample were collected for nucleotide and protein extraction, respectively.

Nucleotide Extraction

Aliquots were pelleted by centrifugation and resuspended in 200 µl of nucleotide extraction buffer (0.1N formic acid and acetonitrile:methanol:water at a 40:40:20), stored on ice for at least 30 mins, and pelleted by centrifugation. Supernatants were retained and vacuum concentrated for ~16 hrs or until samples were dry. Samples were resuspended in 200 µl HPLC-grade dH_2O, vortexed, and pelleted by centrifugation. Supernatant samples were split and stored at -20°C for analysis by either ELISA or LC-MS/MS. ELISA analysis was performed at Cayman Chemical and LC-MS/MS analysis was performed by the Waters Lab in the Department of Microbiology & Molecular Genetics at Michigan State University.

Protein Extraction

Remaining aliquots were pelleted by centrifugation and pellets resuspended in 200 µl of protein extraction buffer (20 mM Tris, pH 6.8 + 1% SDS). Resuspensions were boiled for 10 mins, centrifuged, and supernatants were quantified for total protein using the Bio-Rad DCTM Protein Assay Reagents kit

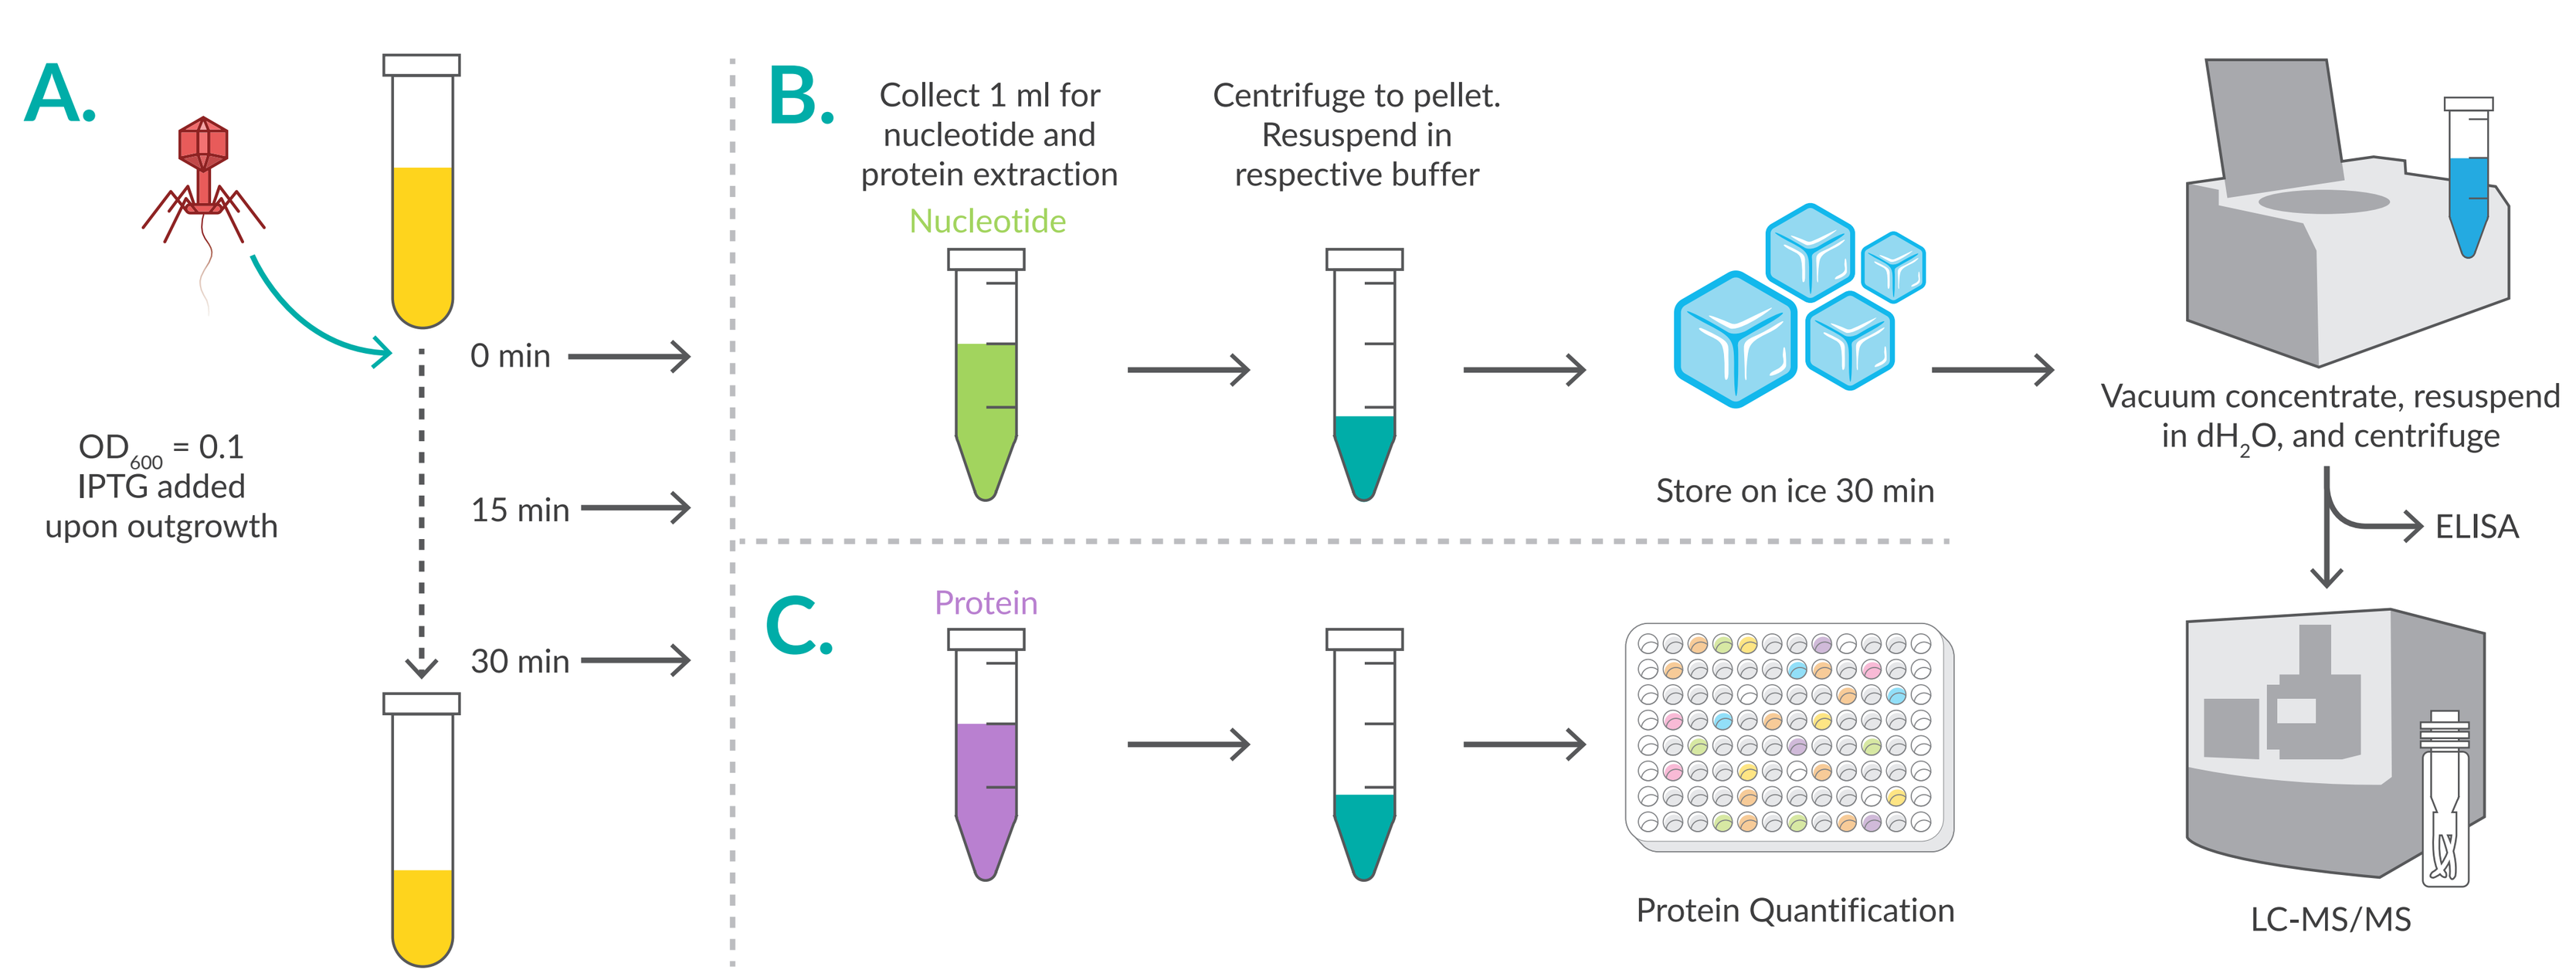
A.
OD600 = 0.1
IPTG added
upon outgrowth
0 min
15 min
30 min
B.
Collect 1 ml for nucleotide and protein extraction
Nucleotide
Centrifuge to pellet. Resuspend in respective buffer
Store on ice 30 min
Vacuum concentrate, resuspend in dH2O, and centrifuge
ELISA
LC-MS/MS
C.
Protein
Protein Quantification

FIGURE 1 – Sample Preparation Workflow for LC-MS/MS and ELISA.
A) Bacterial culture, **B)** Nucleotide extraction for LC-MS/MS and ELISA, **C)** Protein extraction for total protein quantification.

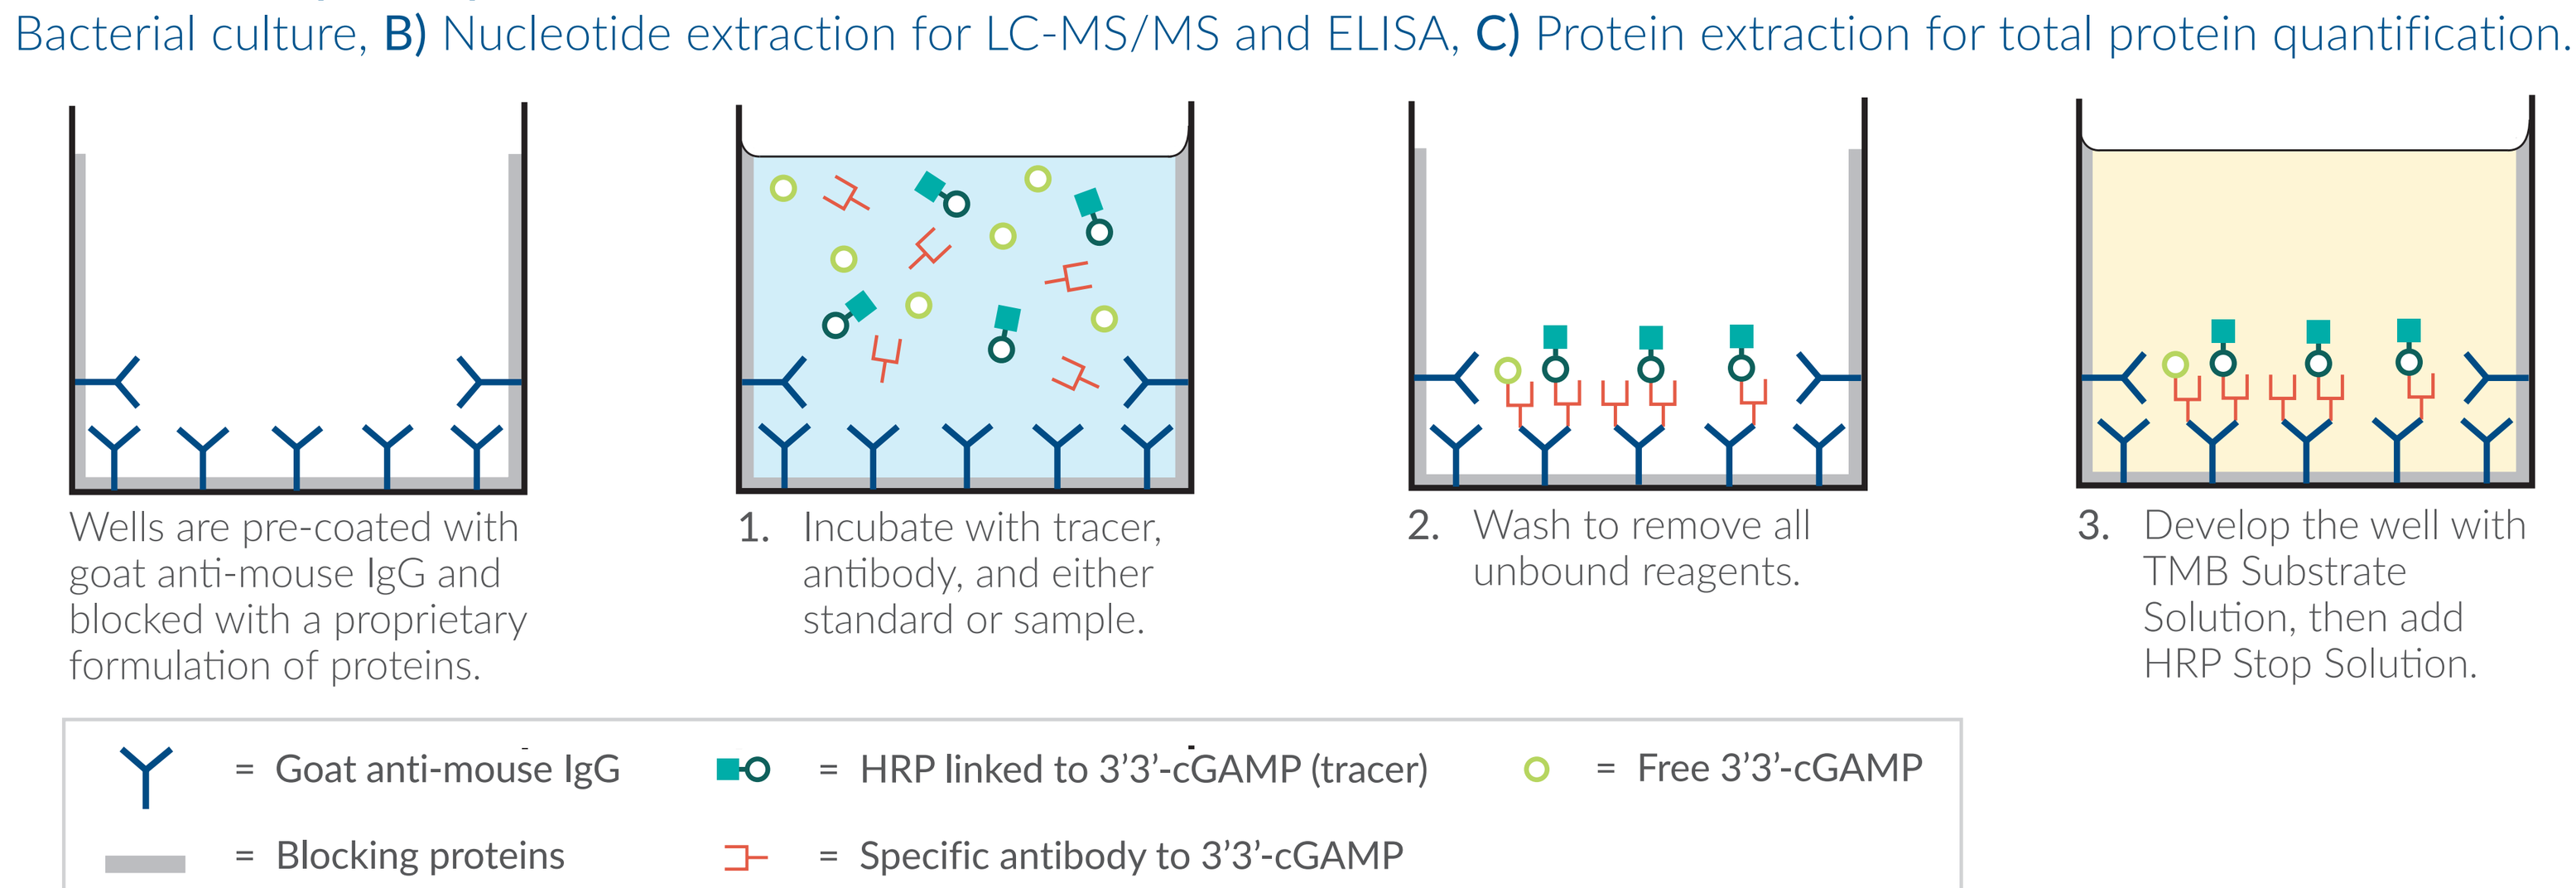
Wells are pre-coated with goat anti-mouse IgG and blocked with a proprietary formulation of proteins.
1. Incubate with tracer, antibody, and either standard or sample.
2. Wash to remove all unbound reagents.
3. Develop the well with TMB Substrate Solution, then add HRP Stop Solution.
= Goat anti-mouse IgG
= HRP linked to 3'3'-cGAMP (tracer)
= Free 3'3'-cGAMP
= Blocking proteins
= Specific antibody to 3'3'-cGAMP

FIGURE 2 – Schematic of the 3'3'-cGAMP ELISA Procedure.

METHODS *(CONTINUED)*

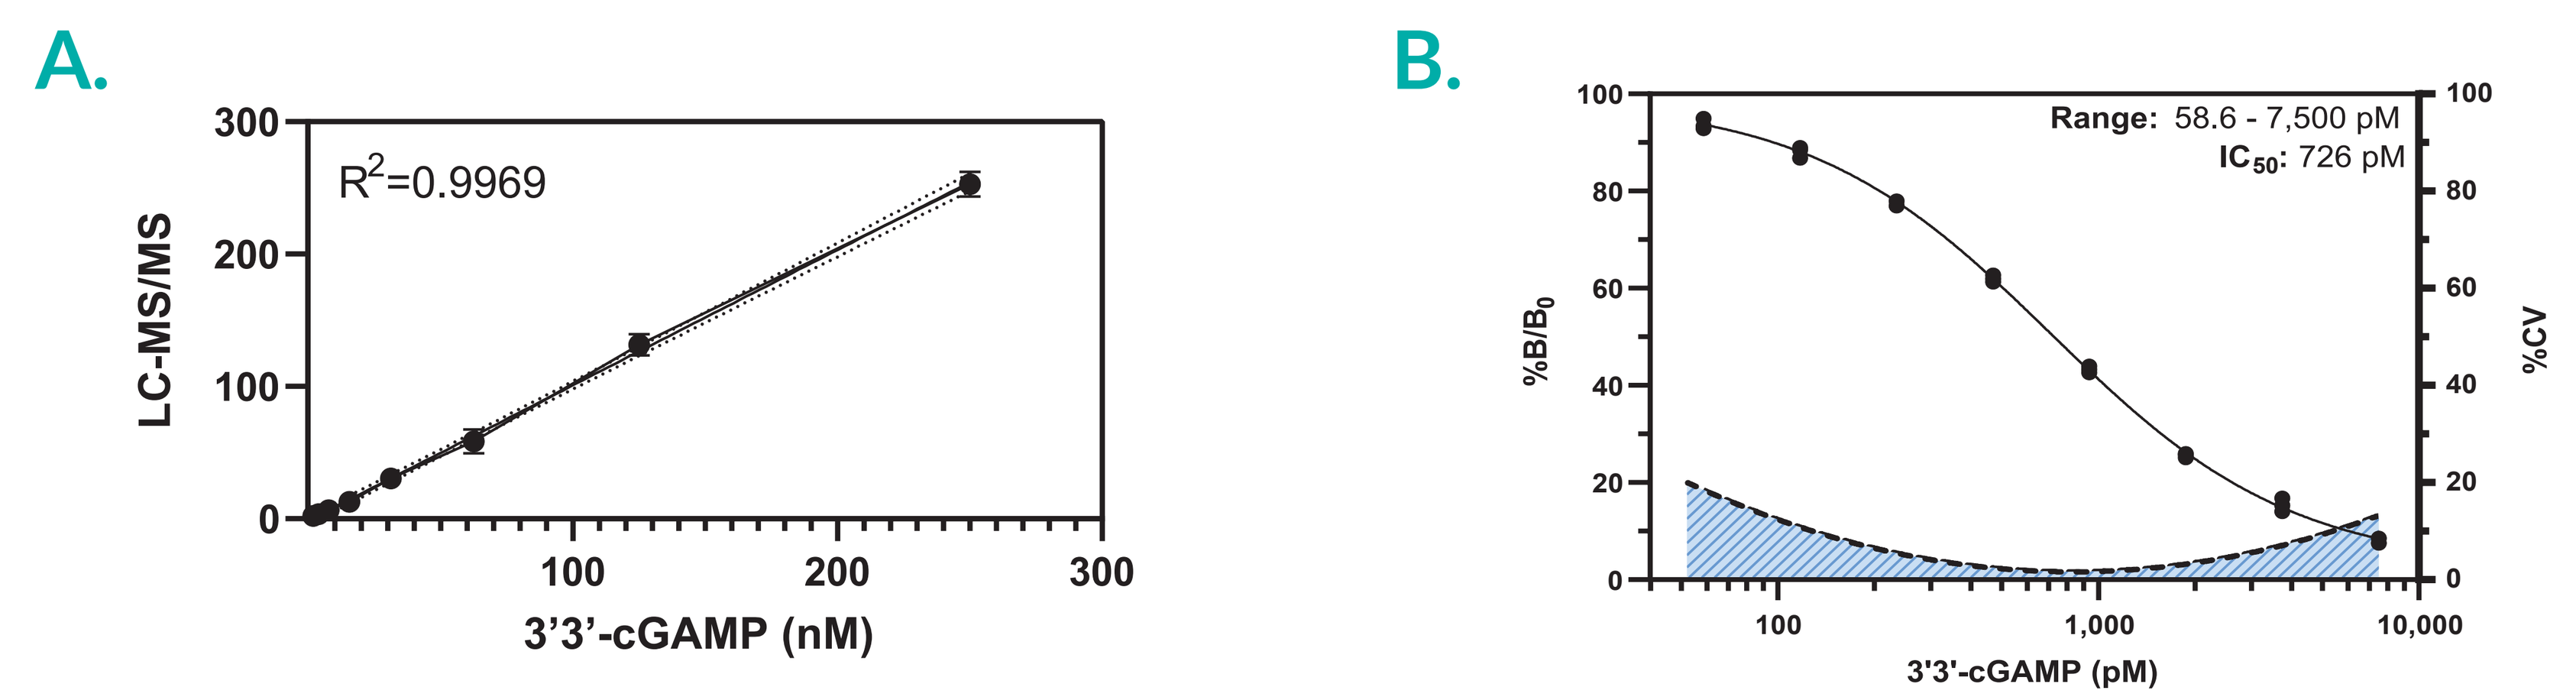
A.
R2=0.9969
LC-MS/MS
3'3'-cGAMP (nM)
B.
Range: 58.6 - 7,500 pM
IC50: 726 pM
%B/B0
%CV
3'3'-cGAMP (pM)

FIGURE 3 – Standard curves for 3'3'-cGAMP quantitation.
A) LC-MS/MS and **B)** ELISA.

ELISA VALIDATION

Specificity

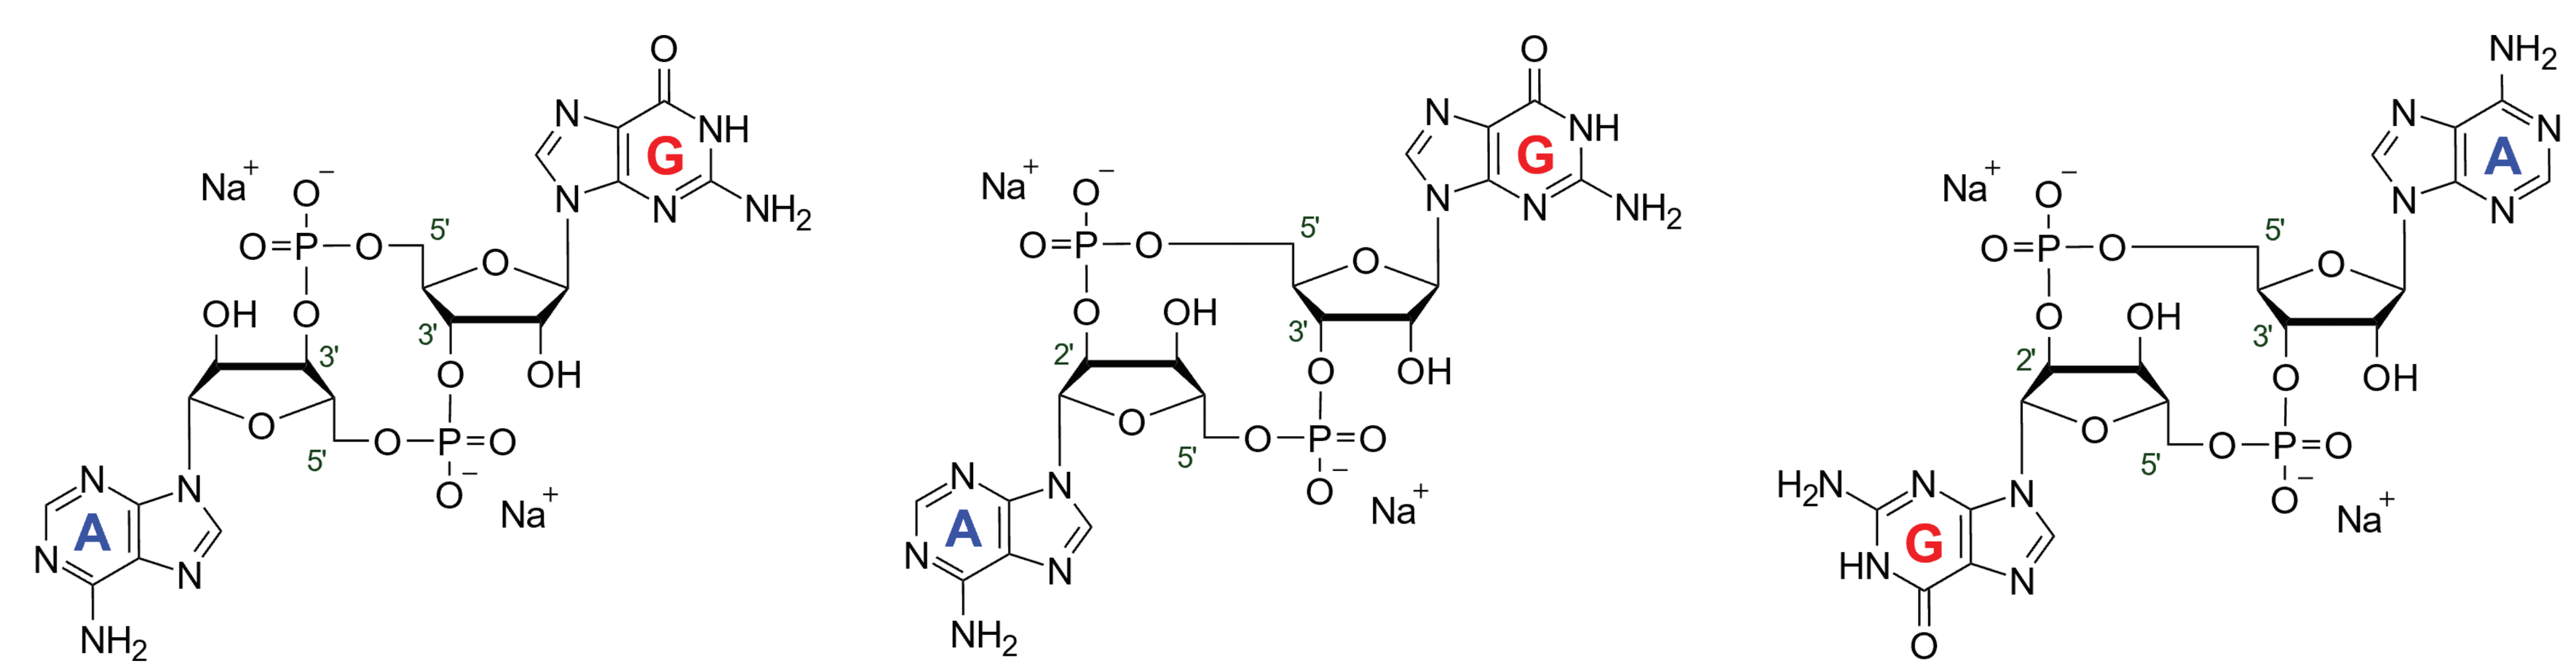

3'3'-cGAMP　　3'2'-cGAMP　　2'3'-cGAMP

Compound	Cross Reactivity (%)
3'3'-cGAMP	100
3'2'-cGAMP	0.018
2'3'-cGAMP	0.006
Cyclic di-GMP	0.004
2'2'-cGAMP	0.002
pApG	0.002

Compound	Cross Reactivity (%)
ATP	<0.001
GTP	<0.001
AMP	<0.001
GMP	<0.001
cAMP	<0.001
cGMP	<0.001

FIGURE 4 – Specificity of the 3'3'-cGAMP ELISA.
Structurally similar and related compounds were tested for cross reactivity in the ELISA. These compounds were serially diluted in assay buffer, evaluated in the ELISA, and compared to the standard curve using the formula below.

$$\%\ Cross\ Reactivity = \left[\frac{50\%\ B/B_0\ value\ for\ the\ 3'3' - cGAMP\ Standard\ Curve}{50\%\ B/B_0\ value\ for\ the\ potential\ cross\ reactant}\right] * 100\%$$

Spike and Recovery

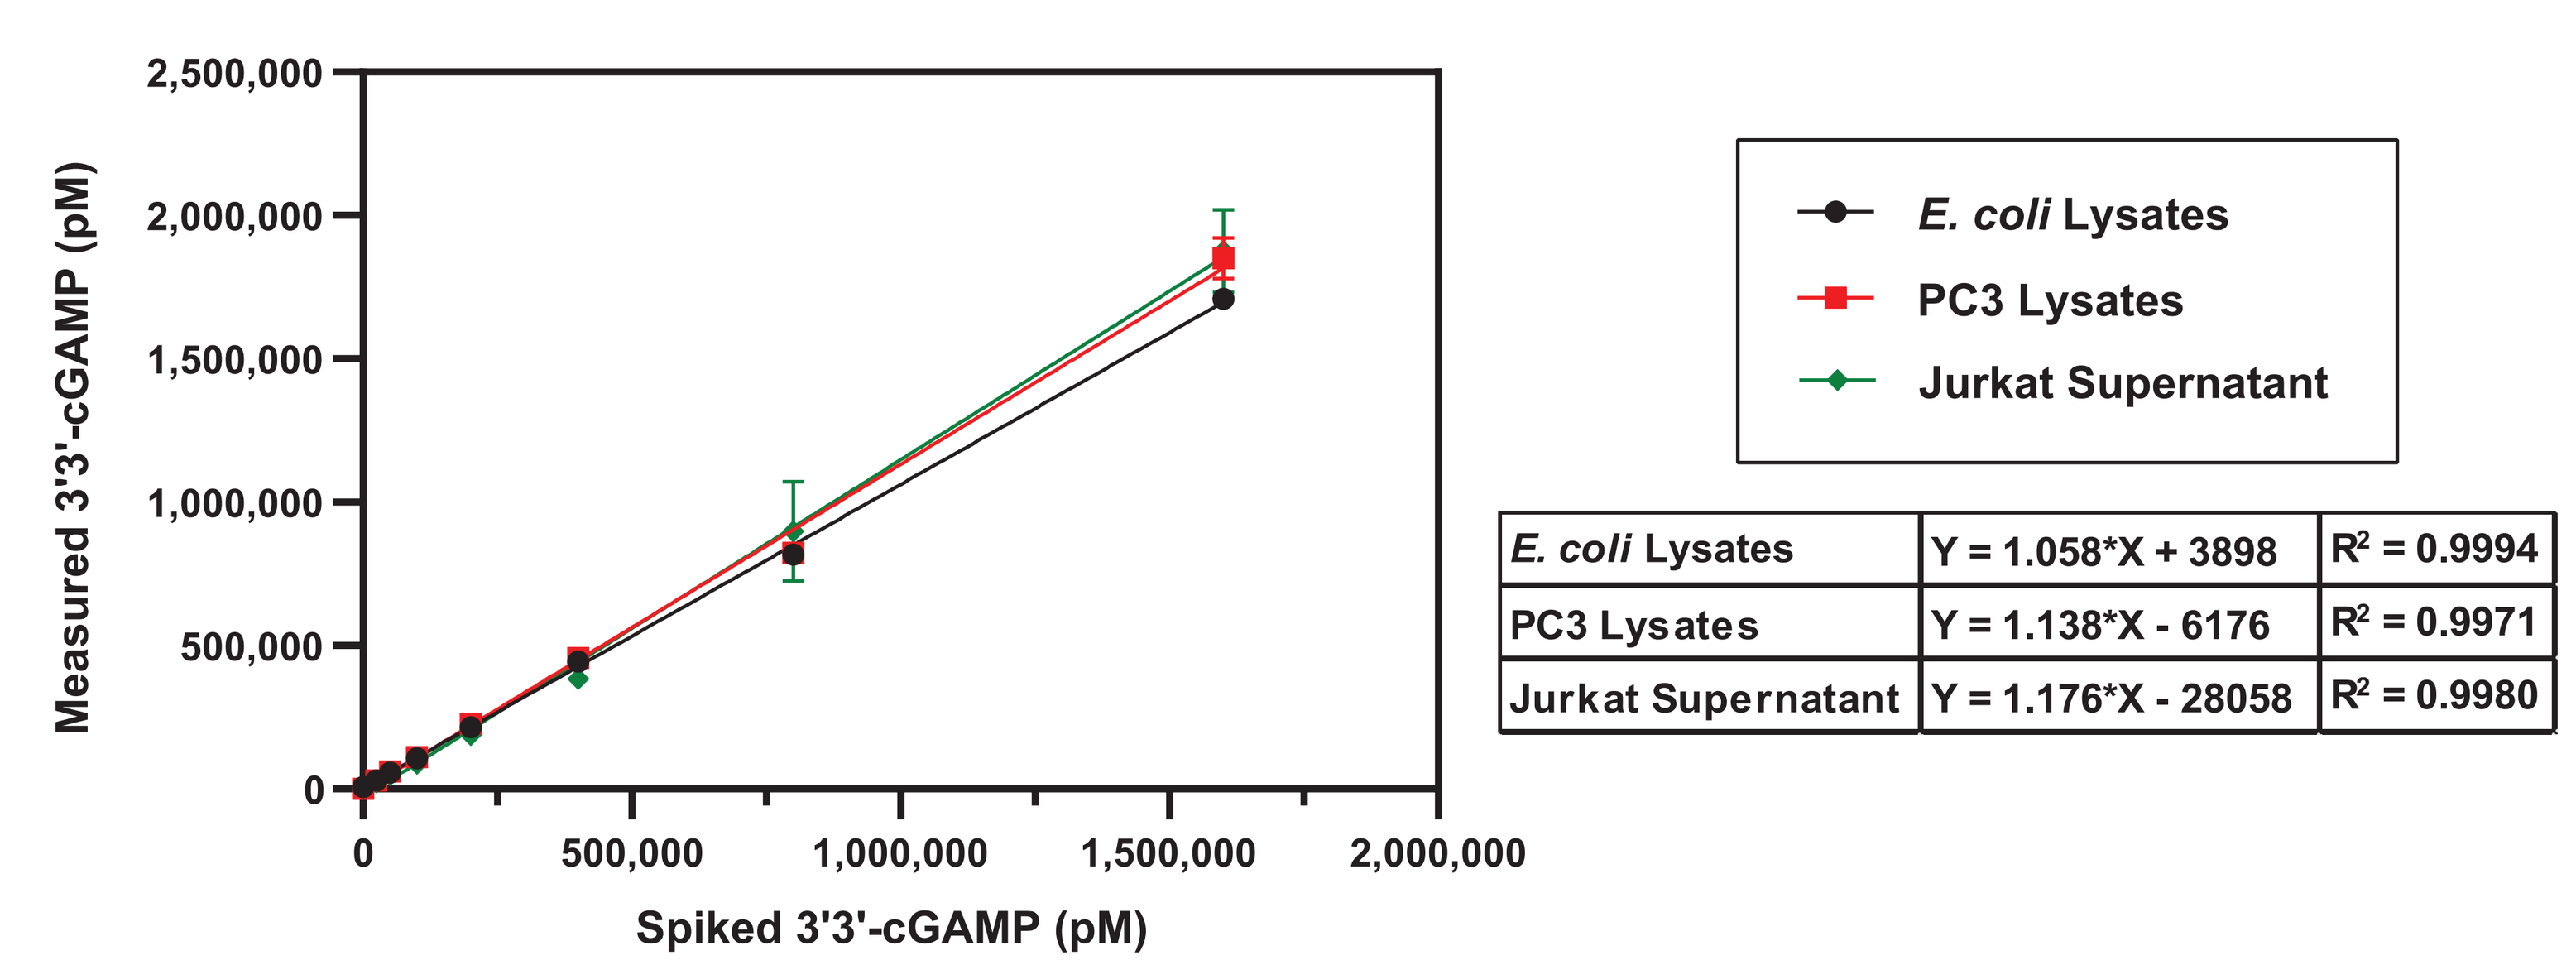
Measured 3'3'-cGAMP (pM)
Spiked 3'3'-cGAMP (pM)
E. coli Lysates
PC3 Lysates
Jurkat Supernatant
E. coli Lysates | Y = 1.058*X + 3898 | R2 = 0.9994
PC3 Lysates | Y = 1.138*X - 6176 | R2 = 0.9971
Jurkat Supernatant | Y = 1.176*X - 28058 | R2 = 0.9980

FIGURE 5 – Spike and recovery in various matrices.
E. coli lysates in B-PER™ (Thermo Scientific), PC3 lysates in M-PER™ (Thermo Scientific), and Jurkat cell supernatant were spiked with different amounts of 3'3'-cGAMP, serially diluted with Immunoassay Buffer C, and evaluated using the 3'3'-cGAMP ELISA Kit. The error bars represent standard deviations from multiple dilutions of each sample.

LC-MS/MS VALIDATION

LC-MS/MS Correlation

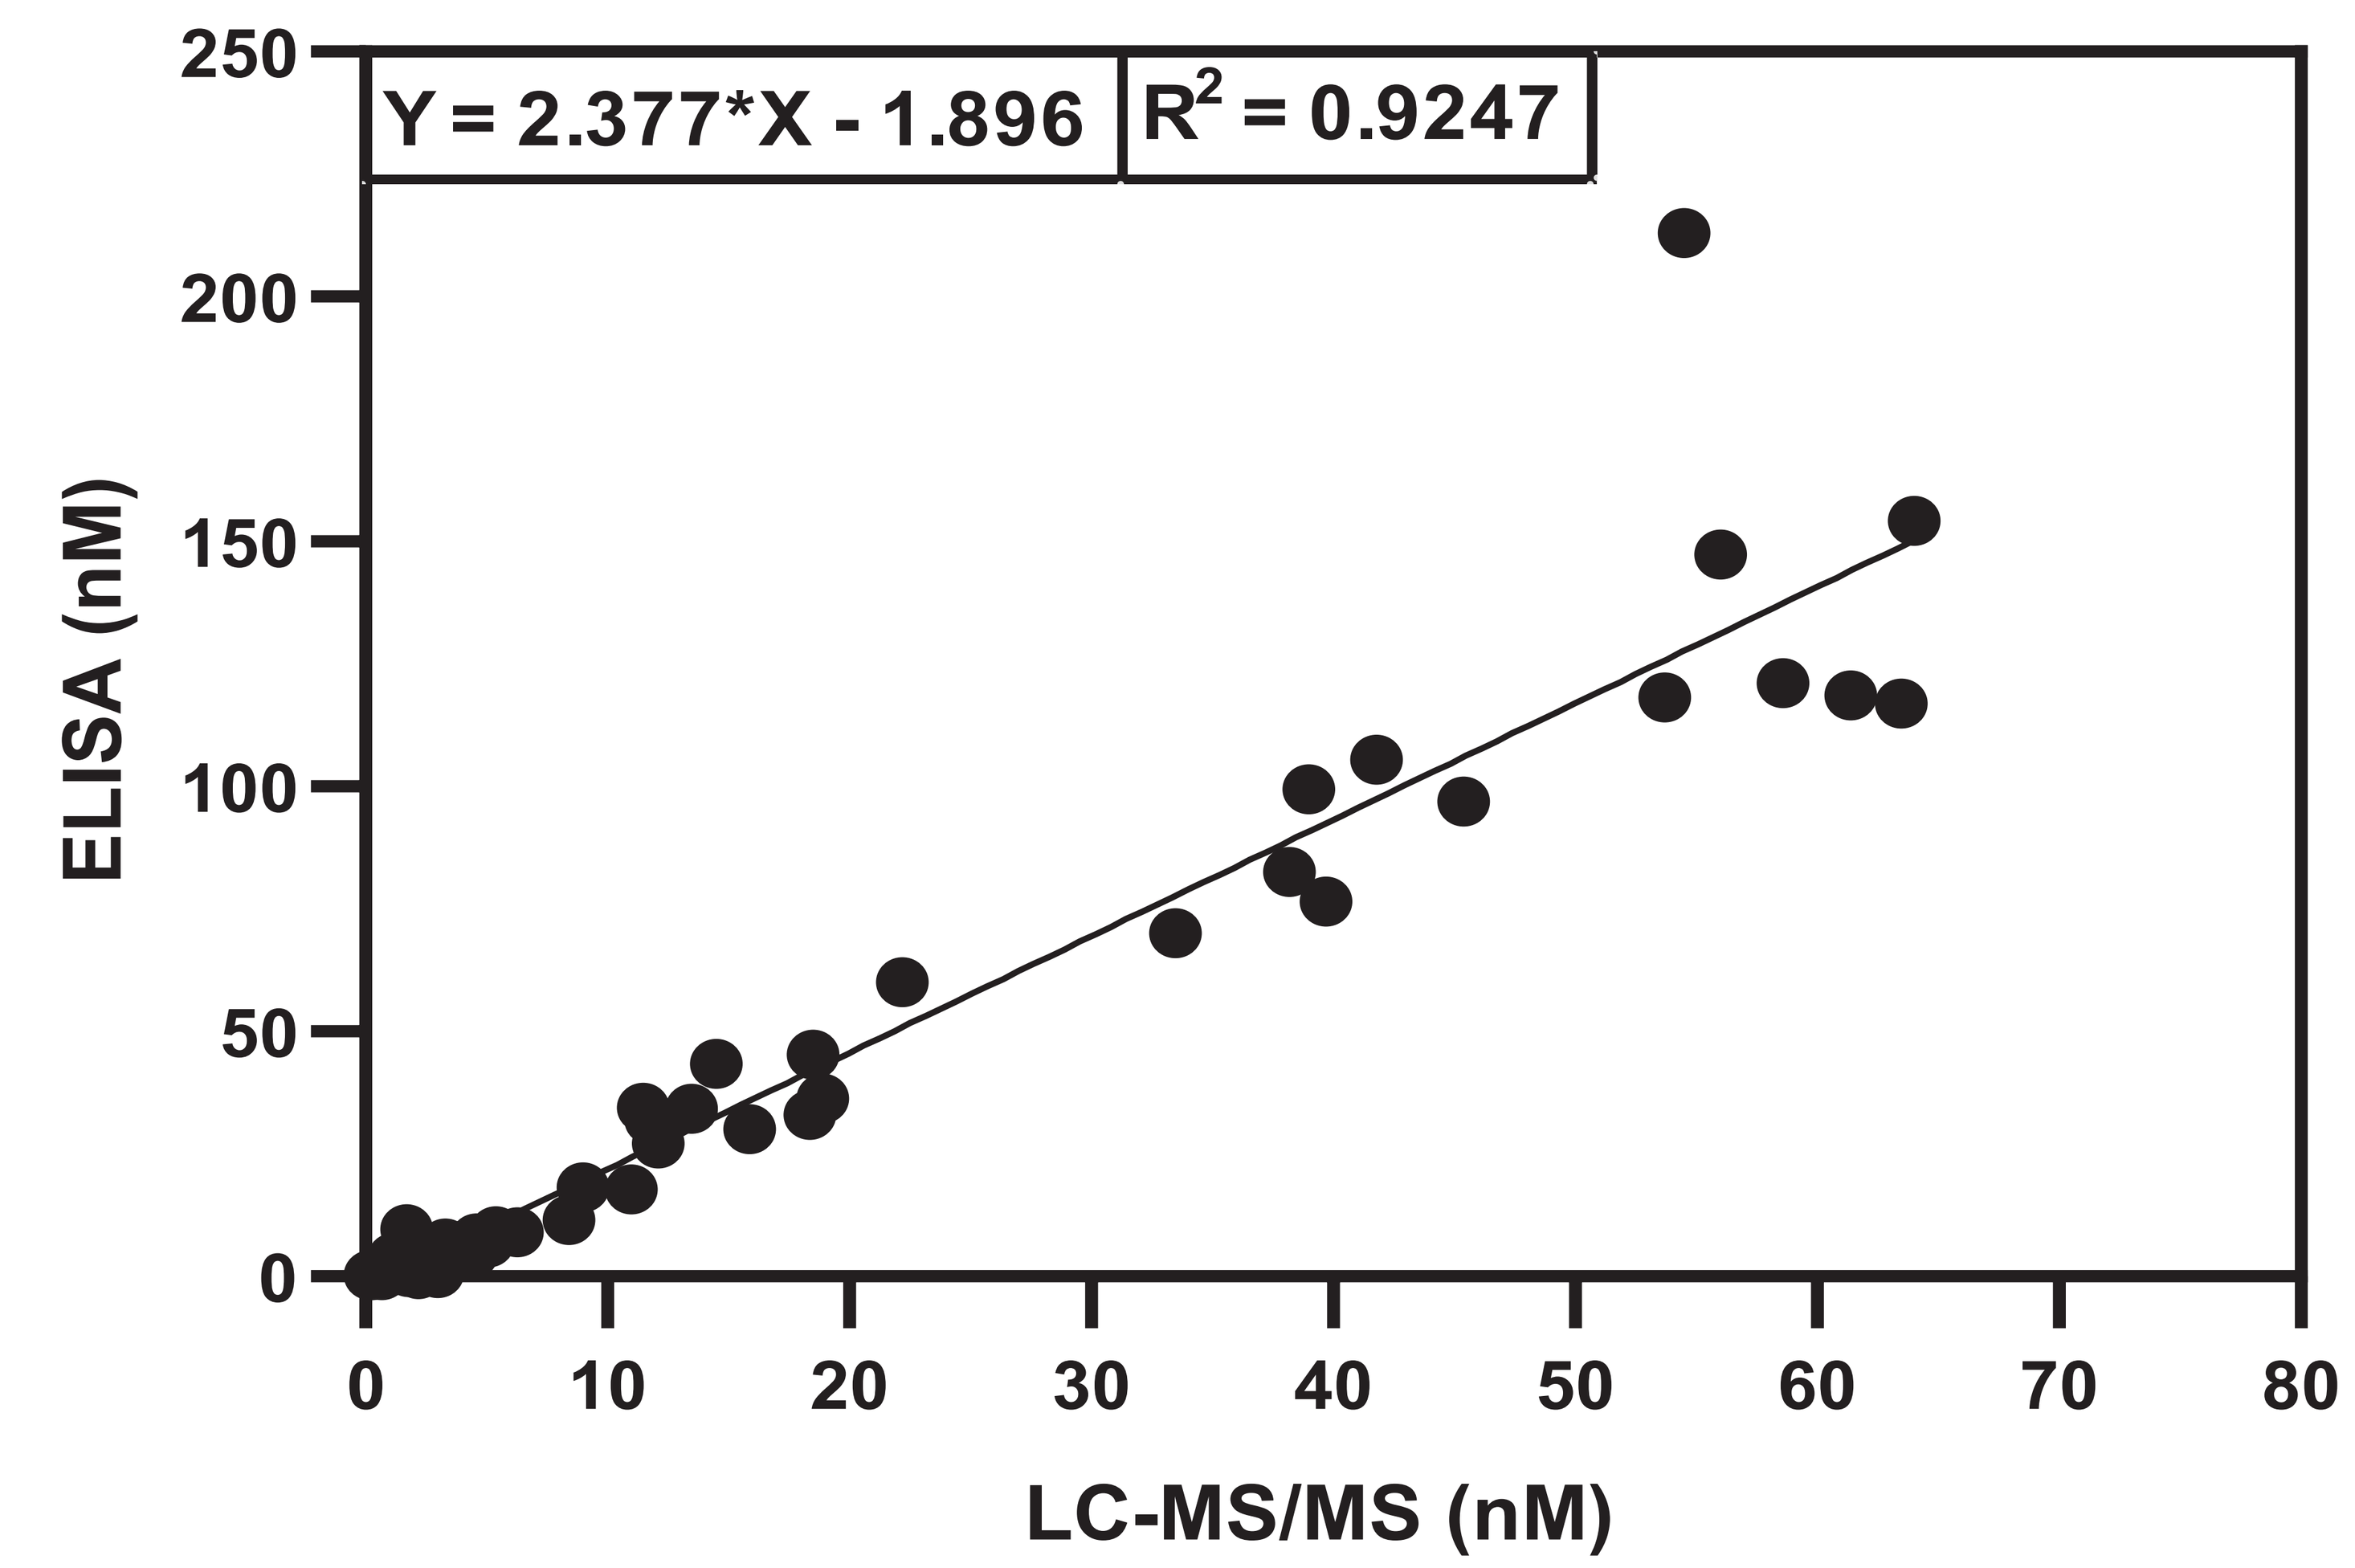
Y = 2.377*X - 1.896
R2 = 0.9247
ELISA (nM)
LC-MS/MS (nM)

FIGURE 6 – Excellent correlation of the 3'3'-cGAMP ELISA to LC-MS/MS.
Transformed and infected *E. coli* samples were generated and measured by LC-MS/MS and compared to values measured by Cayman's 3'3'-cGAMP ELISA Kit. ELISA values were obtained from multiple dilutions of each sample.

CONCLUSIONS

Cayman's novel 3'3'-cGAMP ELISA Kit (**Item No. 502130**) provides an easy-to-use tool for measurement of 3'3'-cGAMP in bacterial and mammalian cell lysates and cell supernatants. Validation of this ELISA has shown outstanding correlation to measurement by LC-MS/MS. This ELISA allows researchers an inexpensive method to study and identify relevant biological pathways.

ACKNOWLEDGEMENTS

A special thanks to Tanya Maxey & Sarah Baitamouni (Validation Department, Cayman Chemical Company, Inc.), for validation of the 3'3'-cGAMP ELISA.

References

1. Davies, B.W., Bogard, R.W., Young, T.S., *et. al.* Coordinated regulation of accessory genetic elements produces cyclic di-nucleotides for *V. cholerae* virulence. *Cell.* **149(2)**, 358–370 (2012). https://doi.org/10.1016/j.cell.2012.01.053
2. Deng, M.J., Tao, J., Chao, E., *et. al.* Novel mechanism for cyclic dinucleotide degradation revealed by structural studies of Vibrio phosphodiesterase V-CGAP3. *J. Mol. Biol.* **430(24)**, 5080–5093 (2018). https://doi.org/10.1016/j.jmb.2018.10.010
3. Wright, T.A., Jiang, L., Park, J.J., Anderson, *et. al.* Second messengers and divergent HD-GYP phosphodiesterases regulate 3',3'-cGAMP signaling. Mol. Microbiol. **113(1)**, 222–236 (2020). https://doi.org/10.1111/mmi.14412
4. Nelson, J.W., Sudarsan, N., Phillips, G.E., *et. al.* Control of bacterial exoelectrogenesis by c-AMP-GMP. *Proc. Natl. Acad. Sci. USA.* **112(17)**, 5389–5394 (2015). https://doi.org/10.1073/pnas.1419264112
5. Wilburn, K.M., Blaylock, J.B., Metcalfe, K.C., *et al.* Development of 3'3'-cyclic GMP-AMP enzyme linked immunoassay reveals phage infection reduced DncV activity. *Isr. J. Chem.* e202200084 (2023).

Check out a use of this ELISA in our application note